Understanding Seizure Clusters from Free-Text Diary Notes

Robert S. Fisher, MD, PhD, Stanford Neurology; **Eyal Bartfeld,** Irody, Inc.; **Joyce Cramer,** Yale

Abstract

Rationale: Patients describe seizure clusters in many ways. Free-text comments in digital epilepsy diaries represent an under-utilized information resource, and give insight (in their own words) into priorities of people with epilepsy. We evaluated patient notes using lexical analysis and diary events to characterize features of seizure clustering, furthering prior observations from diary forms (Fisher RAS, Bartfeld E, Cramer JA. Use of an online epilepsy diary to characterize repetitive seizures. Epilepsy Behav, 2015;47:66-71).

Methods: EpiDiary (www.EpiDiary.com) data includes structured fields for seizure frequency per calendar day, a checkbox for seizure clusters on a given seizure event and a free-text option. For purposes of this analysis, clusters were defined as at least two seizures reported by the patient (including auras or myoclonic jerks) in a calendar day. Lexical analysis was employed to extract comments containing the words: "lots / many / repeat* / frequent* / cluster*" (*=wildcard string, such as "repeatedly"). The authors reviewed every extracted comment to limit results only to text correctly pertaining to clusters. Entries referring to prior clusters or plans for clusters were not included in the analysis. Data were used to characterize repetitive seizures.

Results: 1,839 users, generating a total of 5,955 comments, were flagged by the lexical analysis. Clinician review validated that the comments actually pertained to seizure clusters, as opposed to clusters of migraines, etc., in 2,645 (44.4%) of comments. Comments highlighting clusters did not show a corresponding checkmark in the diary cluster box in 1,560 instances (58.9%). Seizure count was not recorded for 1,192 comments, and 512 comments indicating clusters presented neither a cluster checkmark, nor a seizure count. Extra medicine was taken for clusters by 553 users on 3,818 days, representing 30.1% of all users and 56.5% of those commenting on clusters. Rescue medicines specifically named in a comment were: lorazepam in 99, clonazepam in 84, midazolam in 54, clobazam in 31, rectal diazepam in 29, other diazepam in 18 and clorazepate in 9. Some used more than one rescue medicine. The VNS magnet also was mentioned as a treatment for clusters. Comments often provided potentially useful clinical information, such as "massive ongoing cluster with about 20% apneic events." Interpretations are limited, as not all comments might have been captured. Some subjects used multiple accounts. Words used to identify seizures, e.g., "puffs," might or might not have been about seizures. Occasional comments were copied verbatim from one date to another. Subjects used their own definitions of clusters.

Conclusions: Free-text comments in the digital seizure diary provided additional information about the seizure clusters. More sophisticated lexical and grammatical models will likely improve accuracy of relevant comment identifications in the future.

Methods

- Define cluster as ≥ 2 seizures in a calendar day
- Submit all notes to lexical analysis for words associated with "cluster"
- For example: "lots / many / repeat* / frequent* / cluster*"
- Also highlight mention of rescue medications
- Two reviewers (RF, JC) marked whether the note truly indicated a cluster

Rescue Medicines Named in a Comment

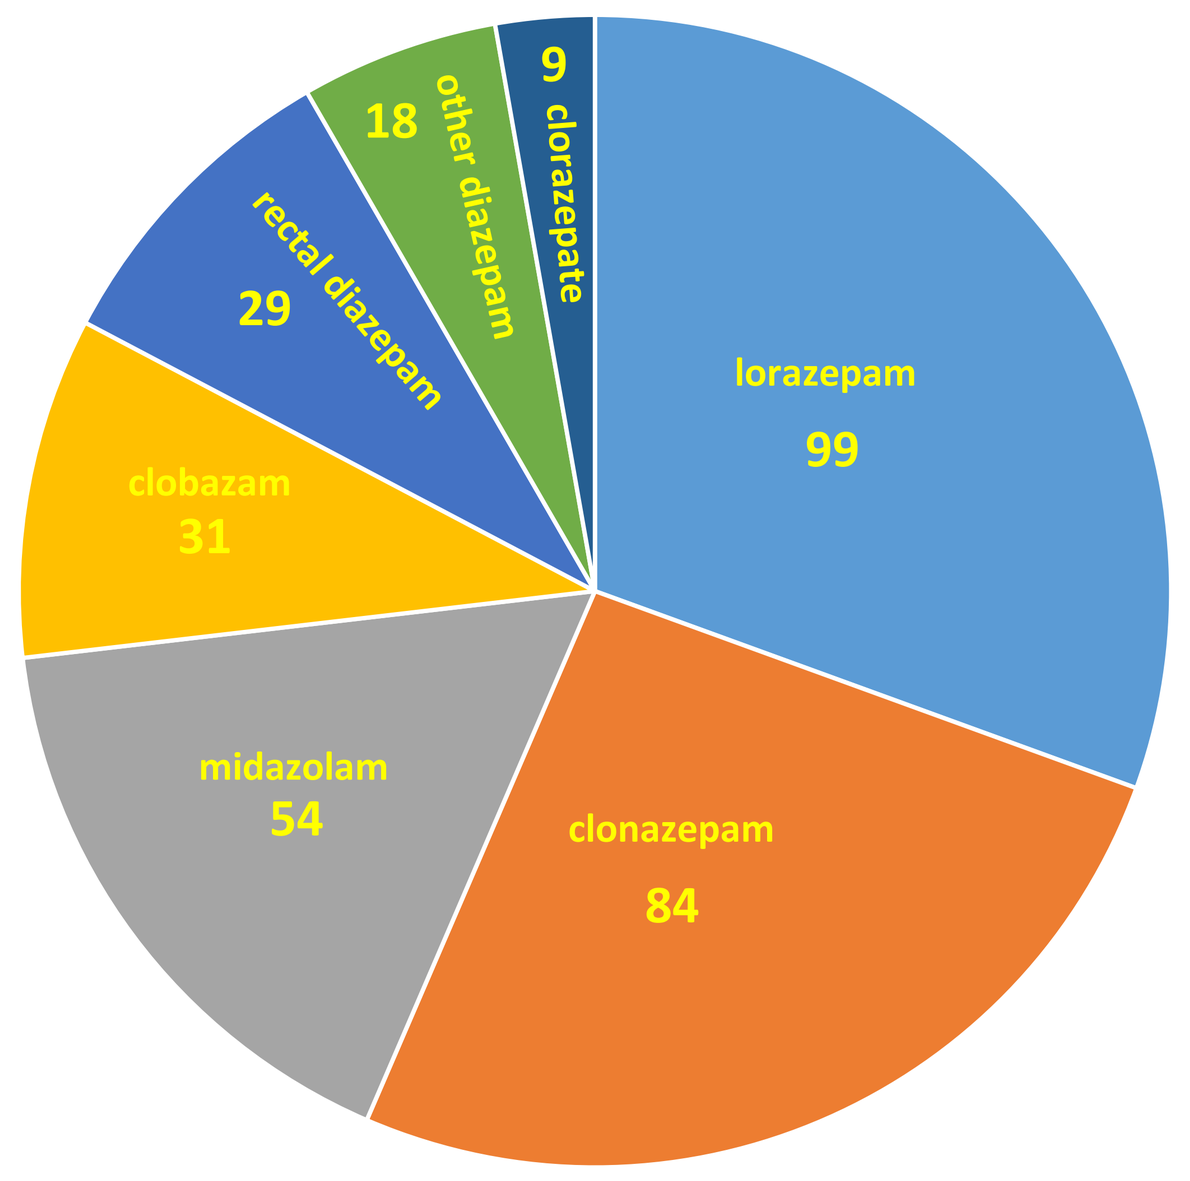
lorazepam
99
clonazepam
84
midazolam
54
clobazam
31
rectal diazepam
29
other diazepam
18
clorazepate
9

Conclusions

- Free-text diary notes are a largely untapped resource
- Important and immediate to the patient, in their own words
- Lexical search rules can extract comments relevant to an issue
- Human validation for 44% of extracted notes about clusters
- Can identify rescue medicines
- 19% of clusters were found only through notes
- Notes contained other clinically relevant information
- This method could apply to many subjects of interest

Questions

- How often do seizure clusters occur?
- Are they reflected in the diary check-box for clusters?
- Are they reflected in free-text notes?
- Is there info on rescue medications?
- Do free-text notes add useful information?

Validation Worksheets

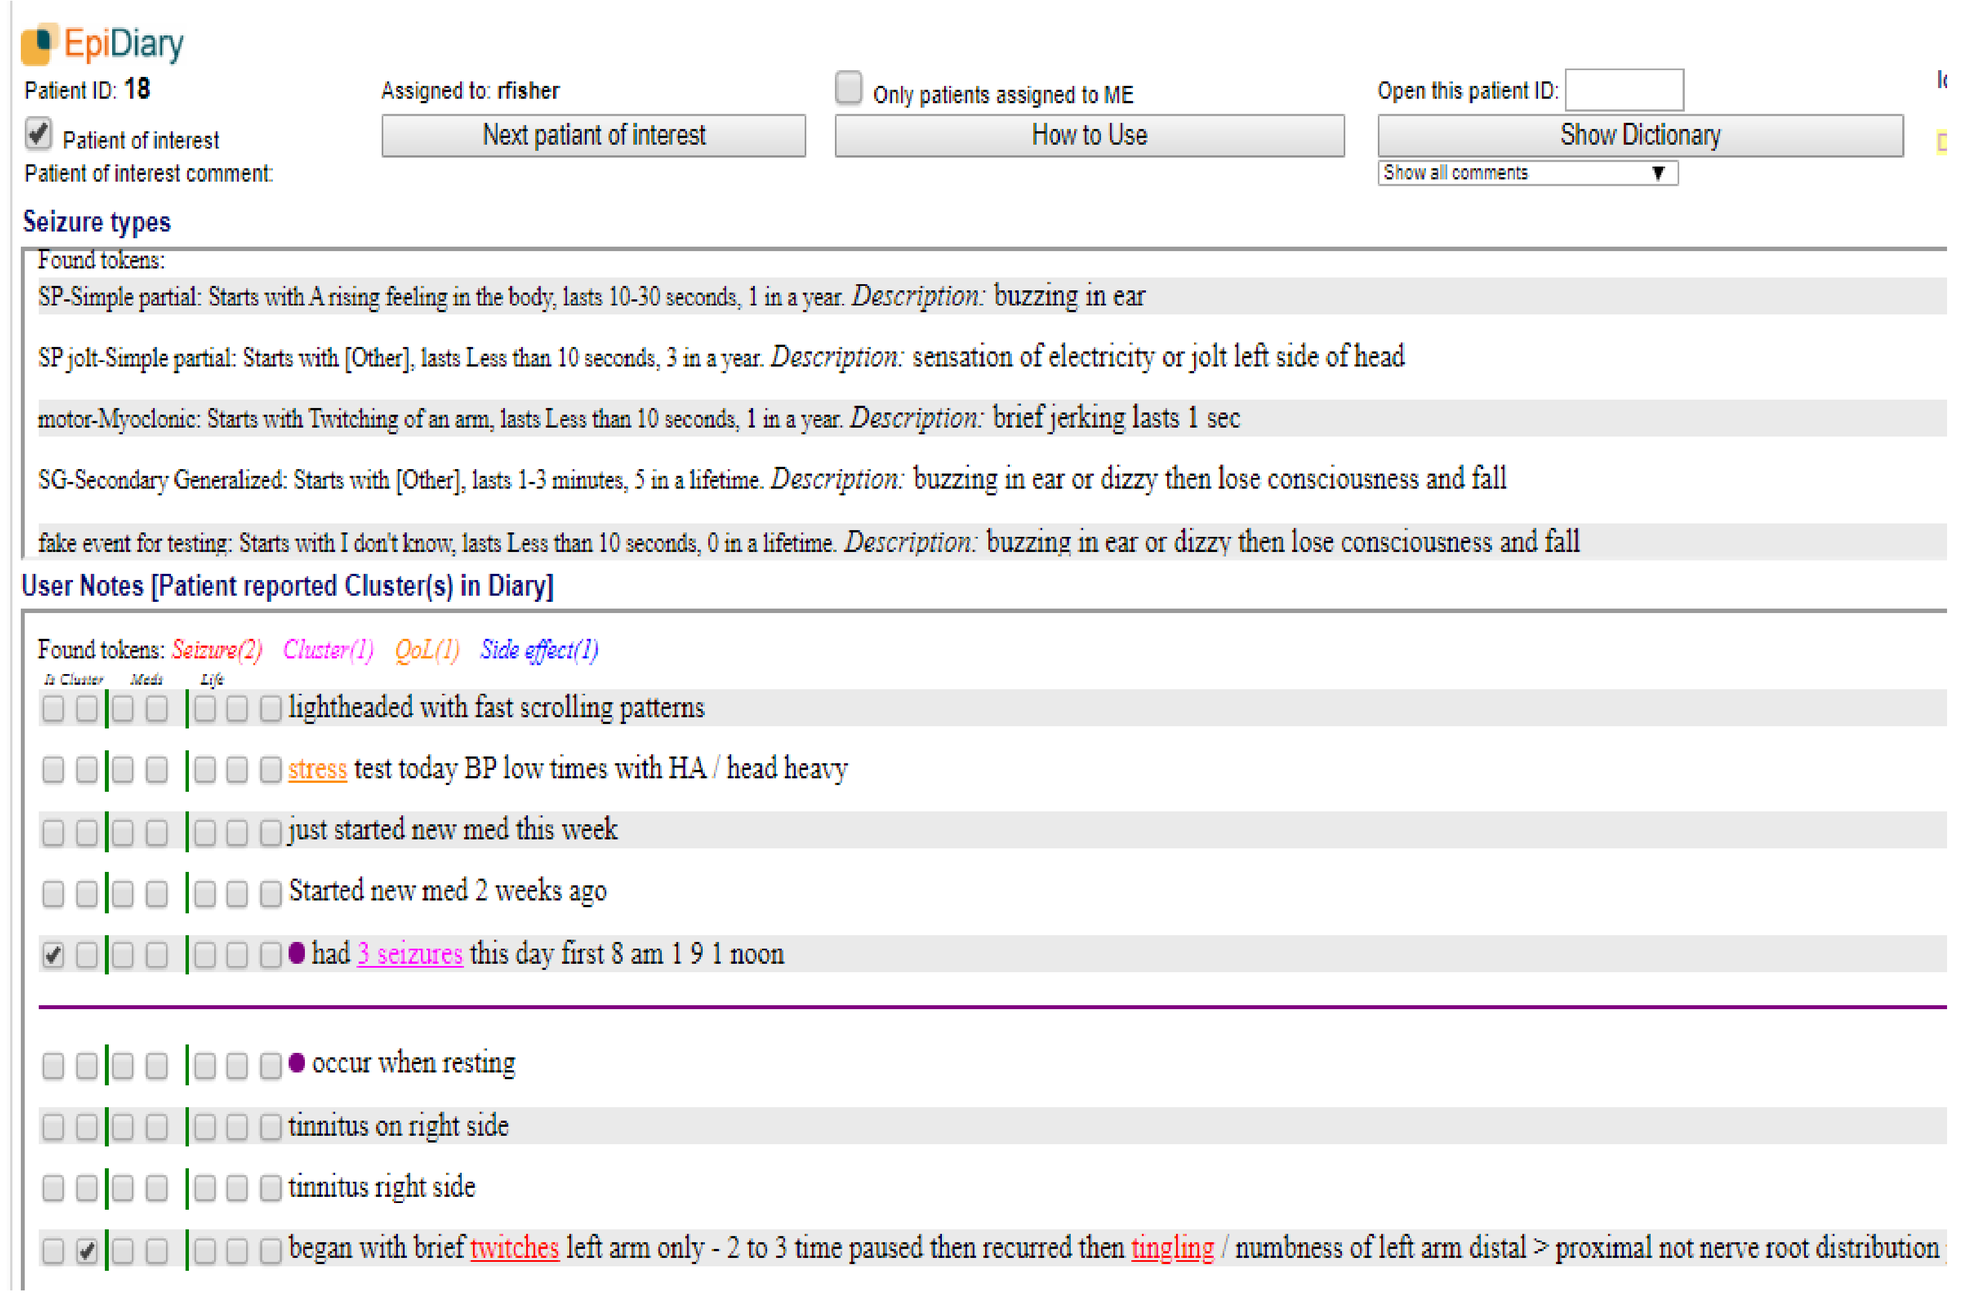
EpiDiary
Patient ID: 18
Assigned to: rfisher
Only patients assigned to ME
Open this patient ID:
Patient of interest
Next patiant of interest
How to Use
Show Dictionary
Patient of interest comment:
Seizure types
Found tokens:
SP-Simple partial: Starts with A rising feeling in the body, lasts 10-30 seconds, 1 in a year. Description: buzzing in ear
SP jolt-Simple partial: Starts with [Other], lasts Less than 10 seconds, 3 in a year. Description: sensation of electricity or jolt left side of head
motor-Myoclonic: Starts with Twitching of an arm, lasts Less than 10 seconds, 1 in a year. Description: brief jerking lasts 1 sec
SG-Secondary Generalized: Starts with [Other], lasts 1-3 minutes, 5 in a lifetime. Description: buzzing in ear or dizzy then lose consciousness and fall
fake event for testing: Starts with I don't know, lasts Less than 10 seconds, 0 in a lifetime. Description: buzzing in ear or dizzy then lose consciousness and fall
User Notes [Patient reported Cluster(s) in Diary]
Found tokens: Seizure(2) Cluster(1) QoL(1) Side effect(1)
lightheaded with fast scrolling patterns
stress test today BP low times with HA / head heavy
just started new med this week
Started new med 2 weeks ago
had 3 seizures this day first 8 am 1 9 1 noon
occur when resting
tinnitus on right side
tinnitus right side
began with brief twitches left arm only - 2 to 3 time paused then recurred then tingling / numbness of left arm distal > proximal not nerve root distribution

Information in Notes Worth Knowing

- Massive ongoing cluster with about 20% apneic events
- My constant question seems to be: HOW can I function in life when just small outings bring about this incessant tiredness?
- Mood swings, crying, aggressive/anger thoughts, depression/suicidal thoughts
- My heart gets very irreg and I get chest pressure and chest pain
- I had hit my head and I had a very bad headache
- Vomited repeatedly (how much can that stomach hold???)
- Started feeling like I was having an aura and pulled over
- Big one while driving home
- Thought about suicide for the first time in a while. Lots of self loathing.

Citations

Haut SR. Seizure clusters: characteristics and treatment. Curr Opin Neurol. 2015;28:143-50.

Asadi-Pooya AA, Nei M, Sharan A, Sperling MR. Seizure clusters in drug-resistant focal epilepsy. Epilepsia 2016;57:e187-90.

Chen B, Choi H, Hirsch LJ, Katz A, Legge A, Wong RA, Jiang A, Kato K, Buchsbaum R, Detyniecki. Prevalence and risk factors of seizure clusters in adult patients with epilepsy. Epilepsy Res. 2017;133:98-102.

Komaragiri A, Detyniecki K, Hirsch LJ. Seizure clusters: A common, understudied and undertreated phenomenon in refractory epilepsy. Epilepsy Behav. 2016;59:83-6.

Fisher RS, Bartfeld E, Cramer JA. Use of an online epilepsy diary to characterize repetitive seizures. Epilepsy Behav. 2015;47:66-71.

Fisher RS, Bartfeld E, Cramer JA. Use of an online epilepsy diary to characterize repetitive seizures. Epilepsy Behav. 2015;47:66-71.

Le S, Shafer PO, Bartfeld E, Fisher RS. An online diary for tracking epilepsy. Epilepsy Behav. 2011 ;22:705-9.

Fisher RS. Tracking epilepsy with an electronic diary. Acta Paediatr. 2010;99:516-8.

Velez M, Fisher RS, Bartlett V, Le S. Tracking generalized tonic-clonic seizures with a wrist accelerometer linked to an online database. Seizure. 2016l;39:13-18.

Fisher RS, Blum DE, DiVentura B, Vannest J, Hixson JD, Moss R, Herman ST, Fureman BE, French JA. Seizure diaries for clinical research and practice: limitations and future prospects. Epilepsy Behav. 2012;24:304-10.

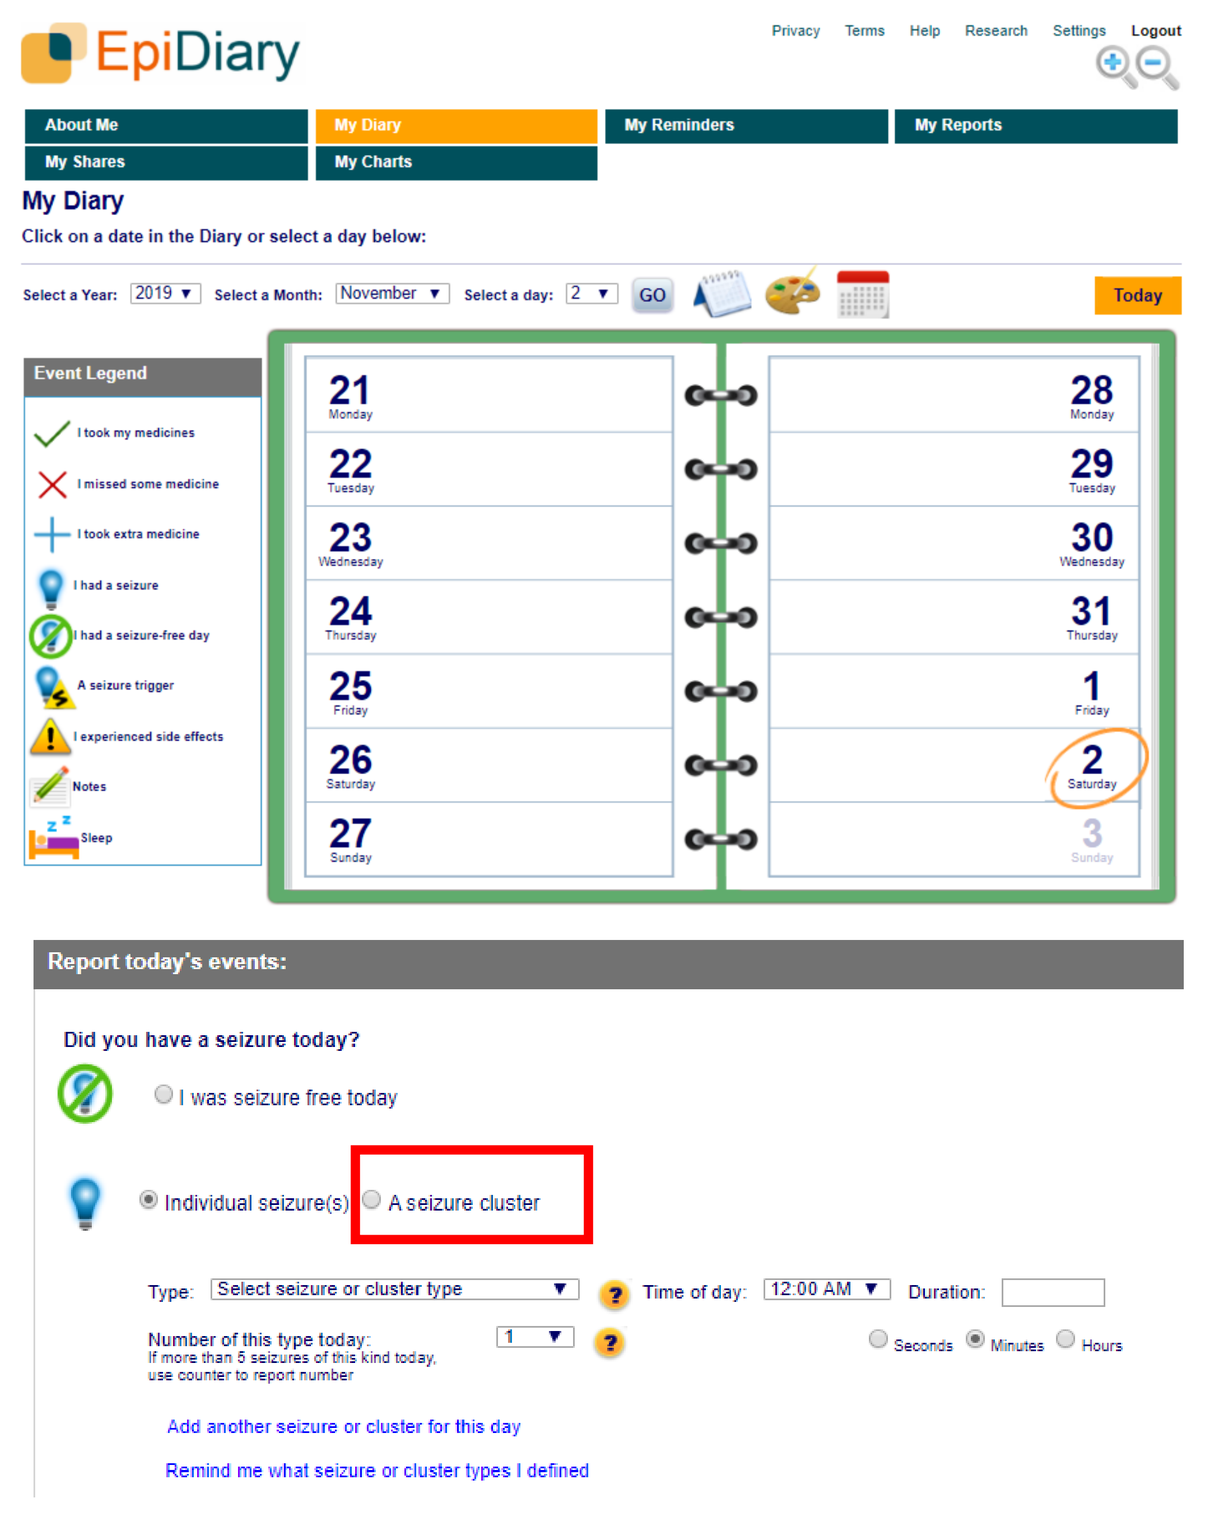
EpiDiary
About Me
My Diary
My Reminders
My Reports
My Shares
My Charts
My Diary
Click on a date in the Diary or select a day below:
Today
Event Legend
Report today's events:
Did you have a seizure today?
I was seizure free today
Individual seizure(s)
A seizure cluster
Add another seizure or cluster for this day
Remind me what seizure or cluster types I defined

EpiDiary

- Structured answers, including "a seizure cluster checkbox
- Also, option for free-text entry

My notes, videos, photos:

A place for entry of any free-text notes for the day

Results

- 1,839 users, with 5,955 comments, were flagged by the lexical analysis.
- Clinician review confirmed clusters for 2645 instances (44.4%).
- Extra medicine was taken for clusters by 553 users on 3,818 days. This was 30.1% of all users and 56.5% of those commenting on clusters.
- Seizure count was not recorded for 1,192 comments.
- Found clusters in 512 comments, with neither a cluster checkmark, nor a seizure count for the day.

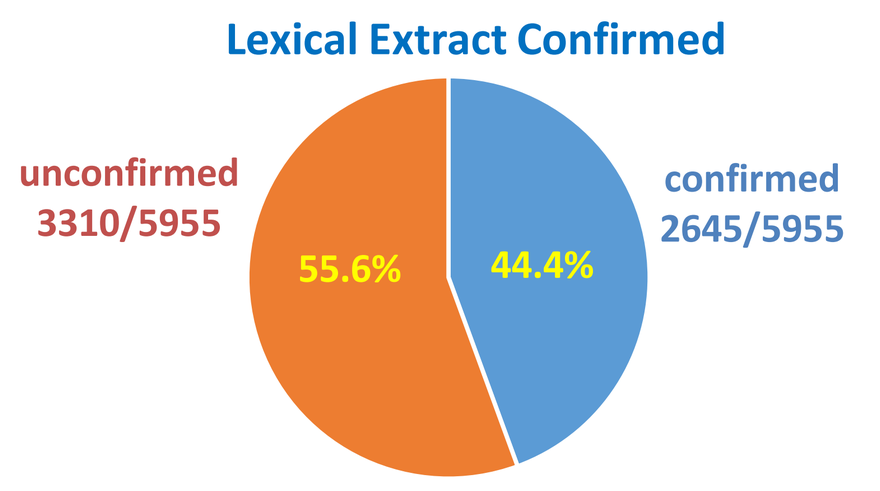
Lexical Extract Confirmed
unconfirmed
3310/5955
55.6%
confirmed
2645/5955
44.4%

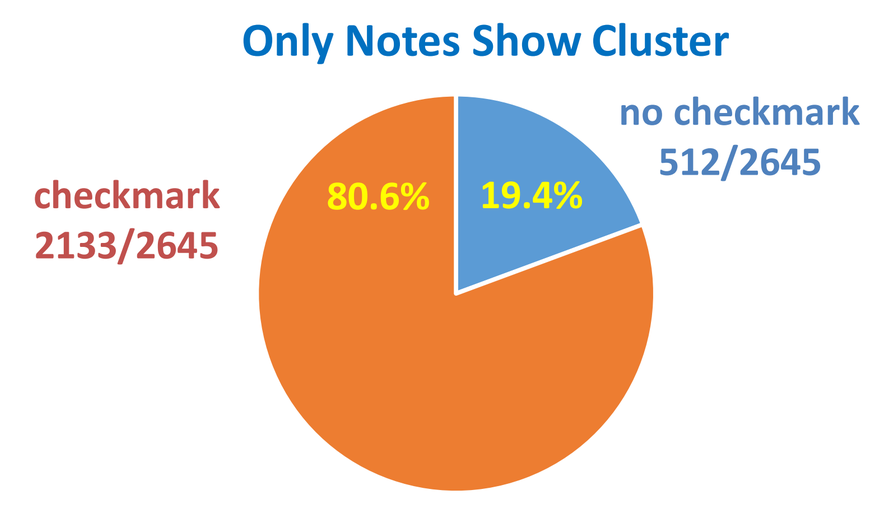
Only Notes Show Cluster
checkmark
2133/2645
80.6%
no checkmark
512/2645
19.4%

Methodological Issues

- Different definitions of seizure clusters
- Words, e.g., "puffs," might or might not have been about seizures
- Occasional comments were copied verbatim from one date to another
- Lexical extractions could be more sensitive and specific

Disclosures

Funding: UCB Pharma and Irody

Disclosures:

- RSF holds stock or options in Irody, Avails Medical, Smart-Monitor, Cerebral Therapeutics, Eysz and consults for Medtronic.
- EB is Founder and President of Irody, Inc.
- JC consults for Eisai, Irody, Sunovion, UCB.